KY1005 a novel anti-OX40L mAb with potential in atopic dermatitis (AD): Results of a Phase 1 study assessing the safety, pharmacokinetics, and T-cell-dependent antibody response (TDAR) in healthy volunteers.

M. Saghari[1] MD, S. Gilbert[2] PhD, M. Yateman[2] PhD, B. Porter-Brown[2] MBBS FFPM, N. Brennan[2] BSc, S. Quaratino[2] MD PhD, R. Wilson[2] PhD, R. Rissmann[1] PhD, MBA van Doorn[3] MD PhD, M. Moerland[1] MD PhD, J. Burggraaf[1,4] MD PhD, P. Gal[1] MD PhD, and J. Powell[2] MBChB FFPM

Introduction

- KY1005 is a human non-depleting monoclonal antibody blocking OX40L interaction with OX40 expressed on activated T cells.
- OX40L is a co-stimulatory molecule, inducibly expressed on antigen-presenting cells.
- This pathway stimulates T_{eff} and suppresses T_{reg} with enhancement of the Th2 response, prolonging the inflammatory response.
- OX40/OX40L expression is upregulated in AD skin.

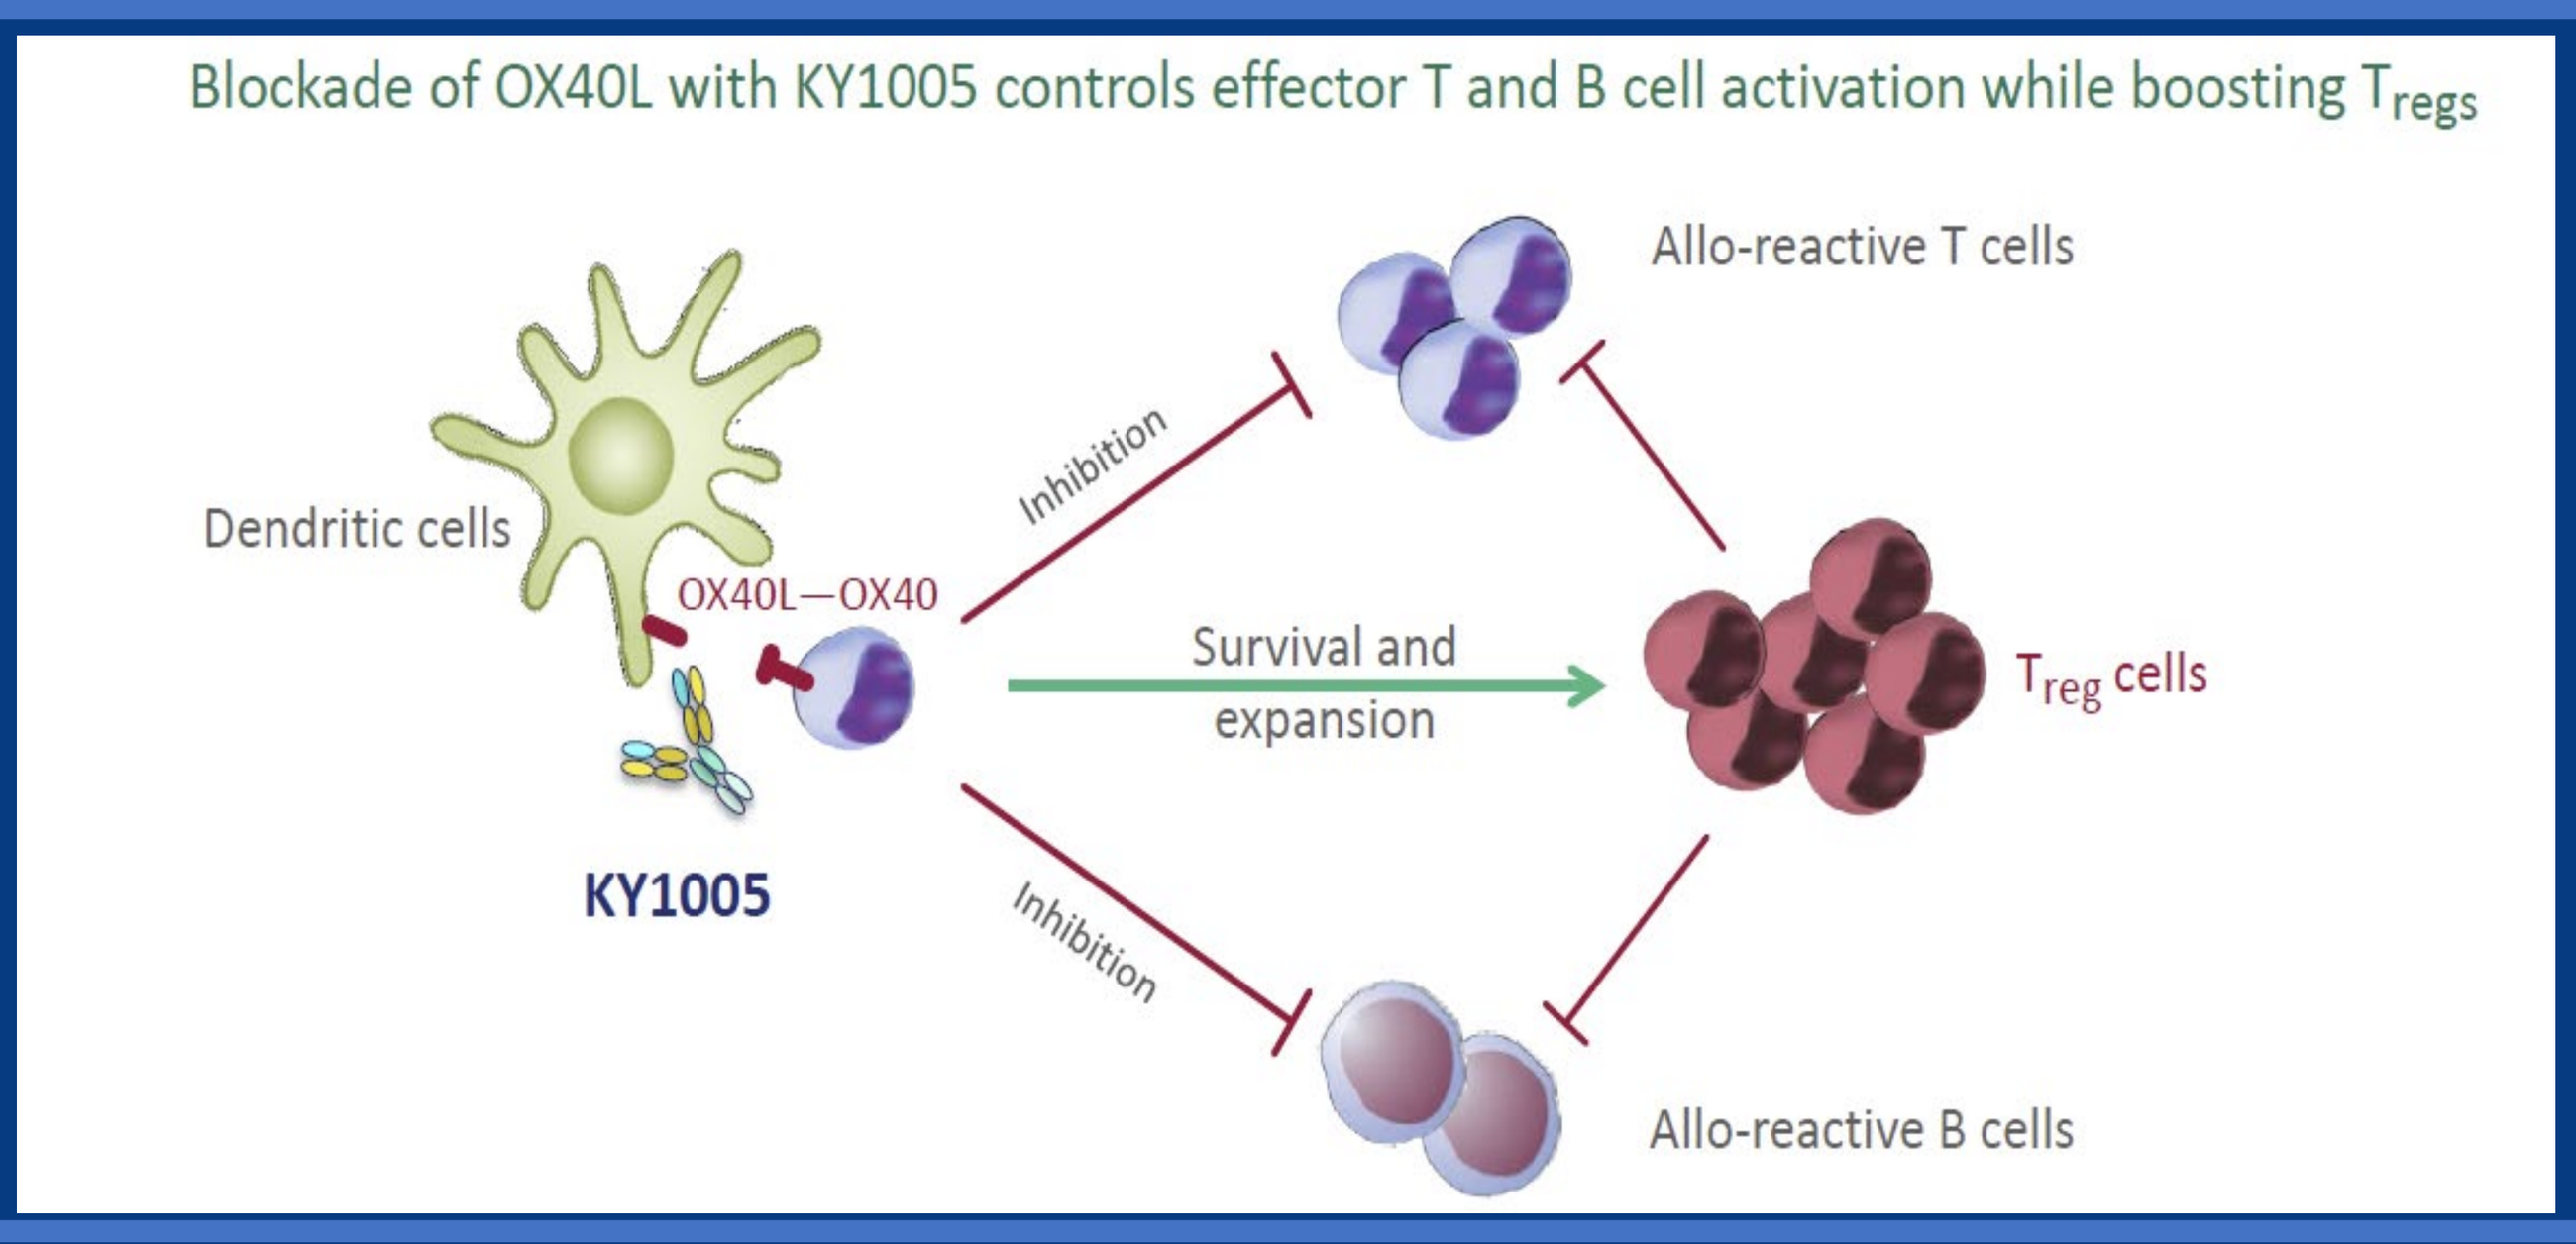
Blockade of OX40L with KY1005 controls effector T and B cell activation while boosting Tregs
Dendritic cells
OX40L—OX40
KY1005
Inhibition
Survival and expansion
Inhibition
Allo-reactive T cells
Treg cells
Allo-reactive B cells

Objective

The primary objective of this Phase I dose escalation study was to evaluate the safety/ tolerability of KY1005 in healthy subjects. Additional objectives included characterisation of the primary TDAR to keyhole limpet hemocyanin (KLH) neoantigen, the recall response to tetanus toxoid (TT), and PK of KY1005.

Materials and Methods

- 64 subjects, recruited across 3 Single-Ascending-Dose cohorts and 5 Multiple-Ascending-Dose (MAD) cohorts (6:2 active vs. placebo ratio).
- MAD cohorts received a loading dose of KY1005 and 2 additional doses (50% of the loading dose) on day 29 and day 57.
- Subjects were followed for up to 16 weeks.
- In the MAD cohorts to assess primary neo-antigen response KLH was used as prior environmental exposure is improbable. Recall antigen response was assesses by TT due to prior exposure via childhood immunisation programs.
- KLH and TT were administered on day 64 by i.m. (left deltoid) administration. The primary neo- and recall antigen T cell dependent antibody response (TDAR) was analysed by serum IgG and IgM, 21 days later on day 85
- KY1005 concentrations were assessed throughout.

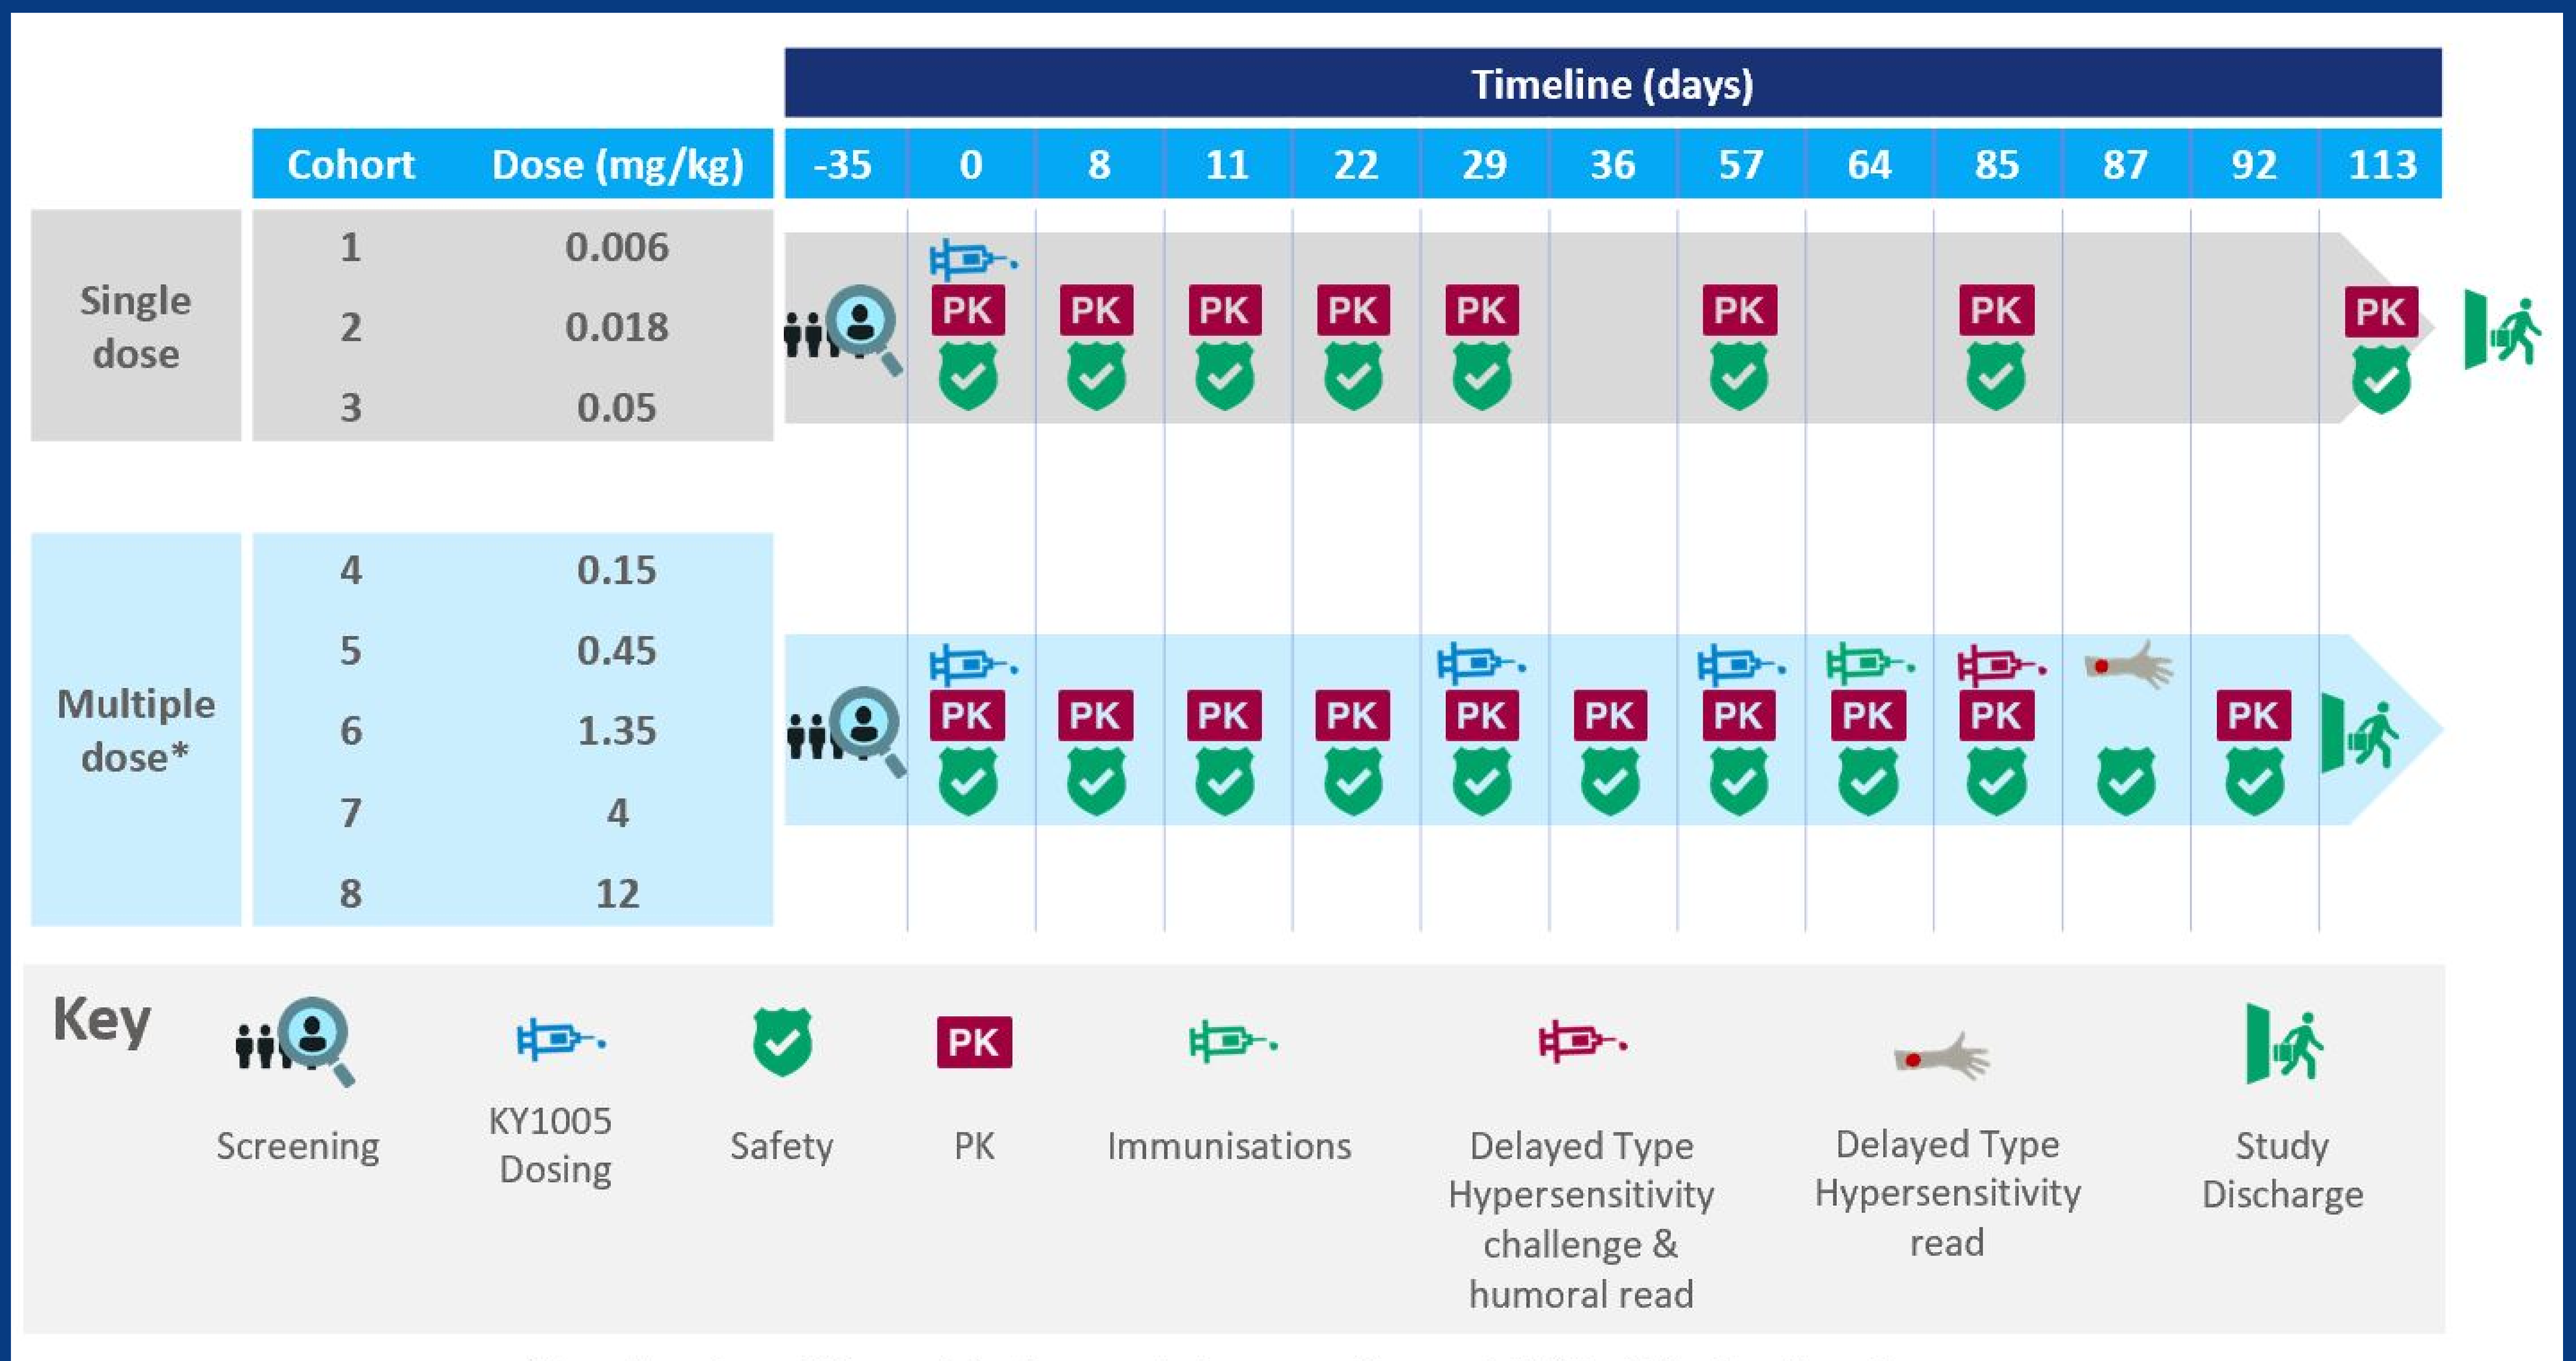
Timeline (days)
Cohort
Dose (mg/kg)
-35
0
8
11
22
29
36
57
64
85
87
92
113
Single dose
1 0.006
2 0.018
3 0.05
Multiple dose*
4 0.15
5 0.45
6 1.35
7 4
8 12
PK
Key
Screening
KY1005 Dosing
Safety
PK
Immunisations
Delayed Type Hypersensitivity challenge & humoral read
Delayed Type Hypersensitivity read
Study Discharge

*Loading dose followed by two maintenance doses at 50% of the loading dose

Results

- No deaths, SAEs, severe hypersensitivity or severe injection site reactions occurred.
- One hypersensitivity reaction localised to the throat/mouth occurred during the first infusion at the top dose, 12mg/kg. This resolved without intervention.
- Suppression of the TDAR against a neo-antigen (KLH subunit) was observed from doses 0.45 mg/kg and above (Cohorts 5 to 8). The suppression in the IgG response was more marked than the IgM response. No clear effect was observed after recall antigen exposure, although, in general, anti-TT IgM antibody titres were numerically smaller in KY1005-treated subjects compared to placebo.
- Half-life ranged between 20 to 42 days with no dose-related trend.

Effect of KY1005 on anti KLH antibody production 21 days following immunisation

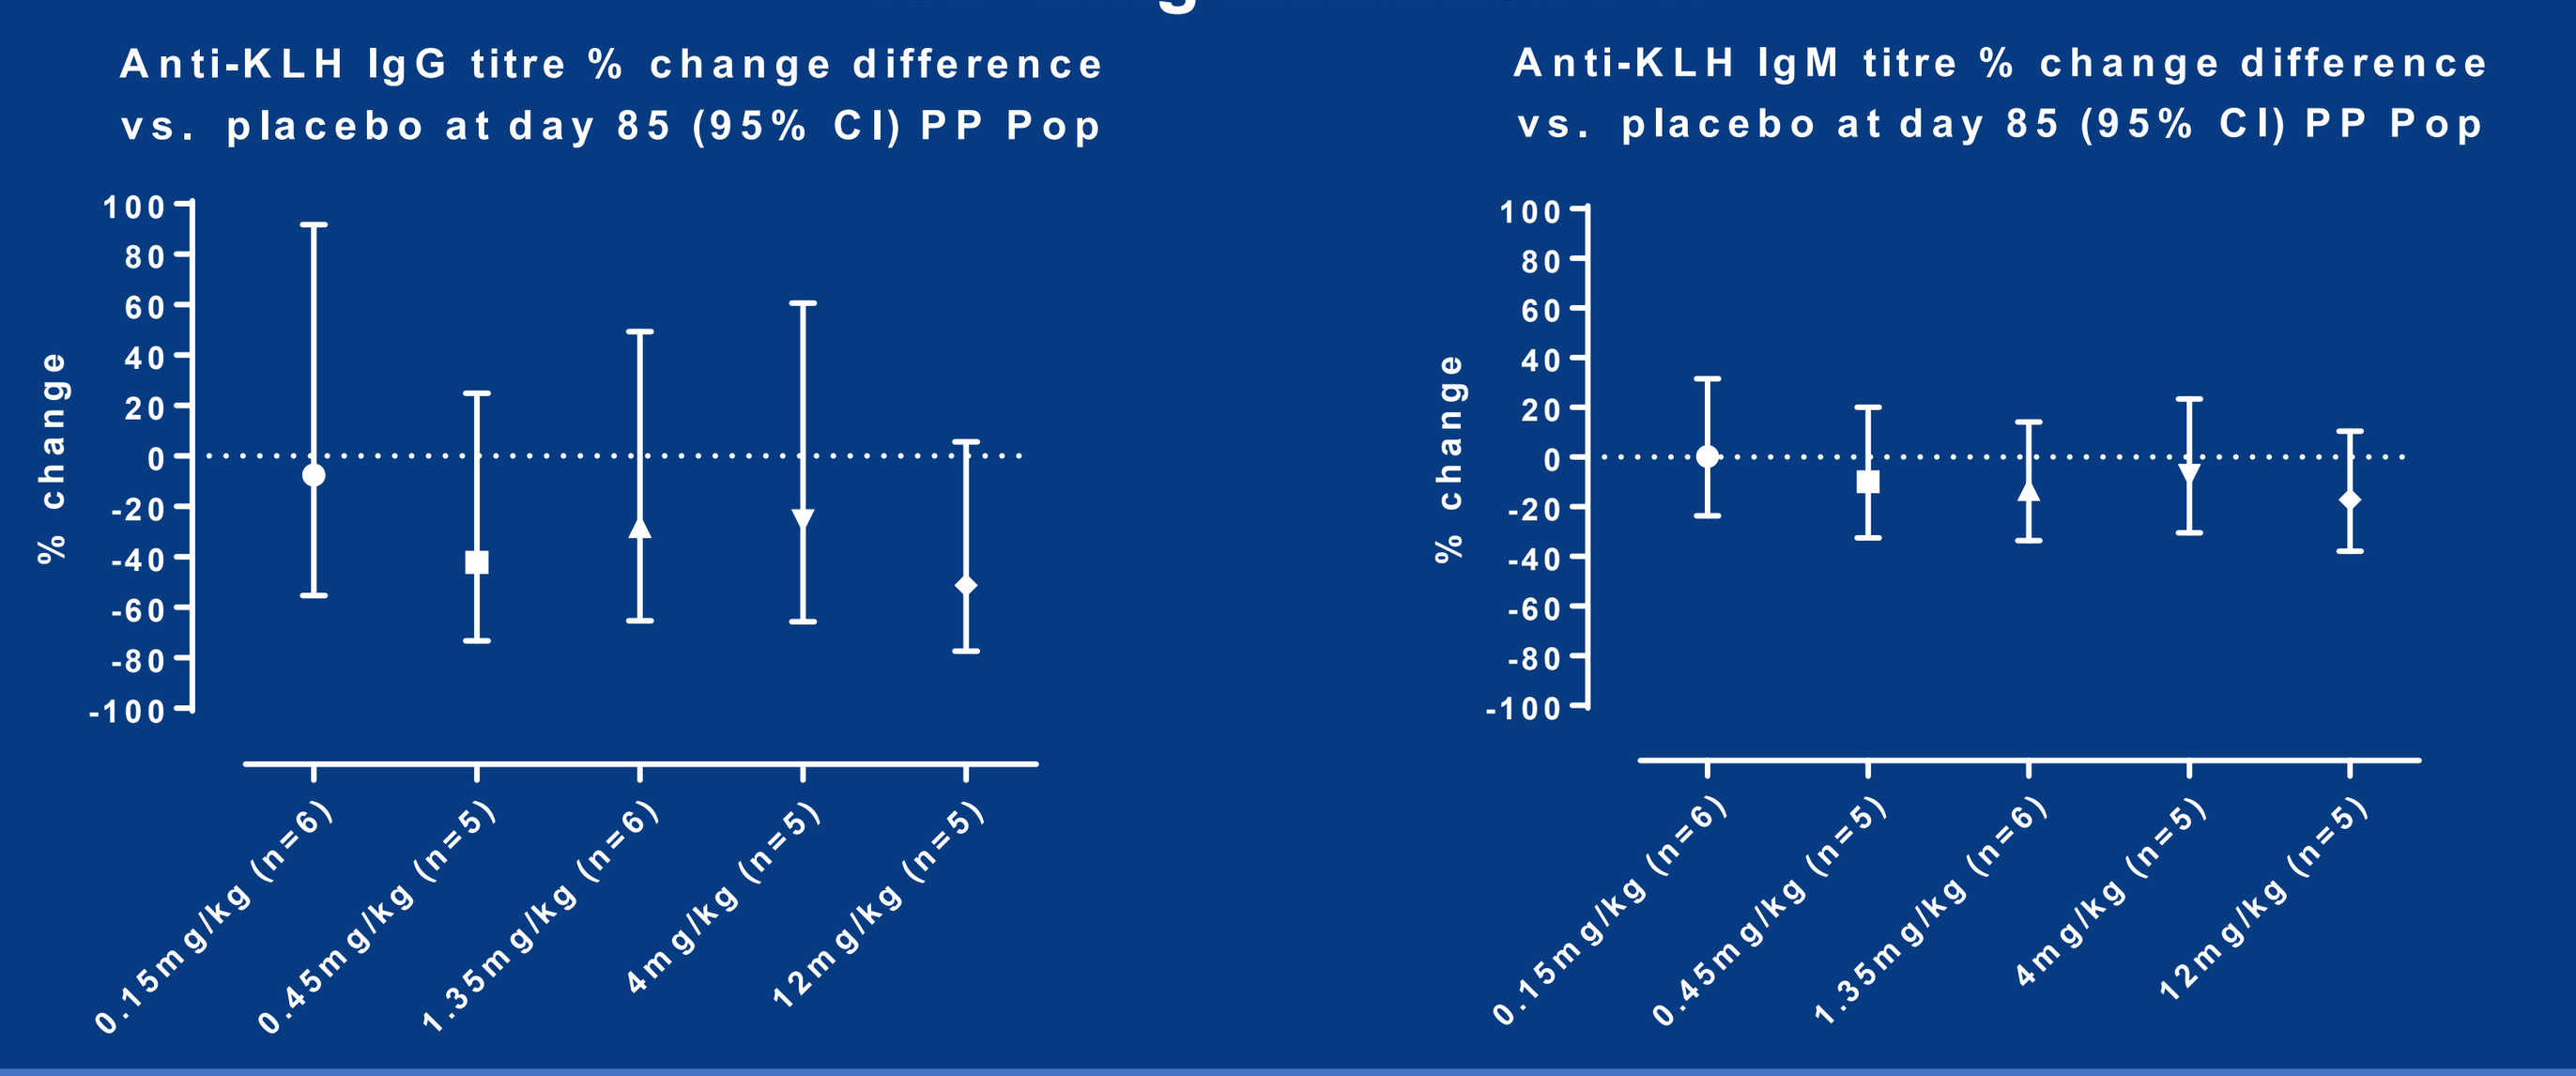
Anti-KLH IgG titre % change difference vs. placebo at day 85 (95% CI) PP Pop
Anti-KLH IgM titre % change difference vs. placebo at day 85 (95% CI) PP Pop
% change
100
80
60
40
20
0
-20
-40
-60
-80
-100
0.15mg/kg (n=6)
0.45mg/kg (n=5)
1.35mg/kg (n=6)
4mg/kg (n=5)
12mg/kg (n=5)

Serum KY1005 (ng/mL) over time by dose group

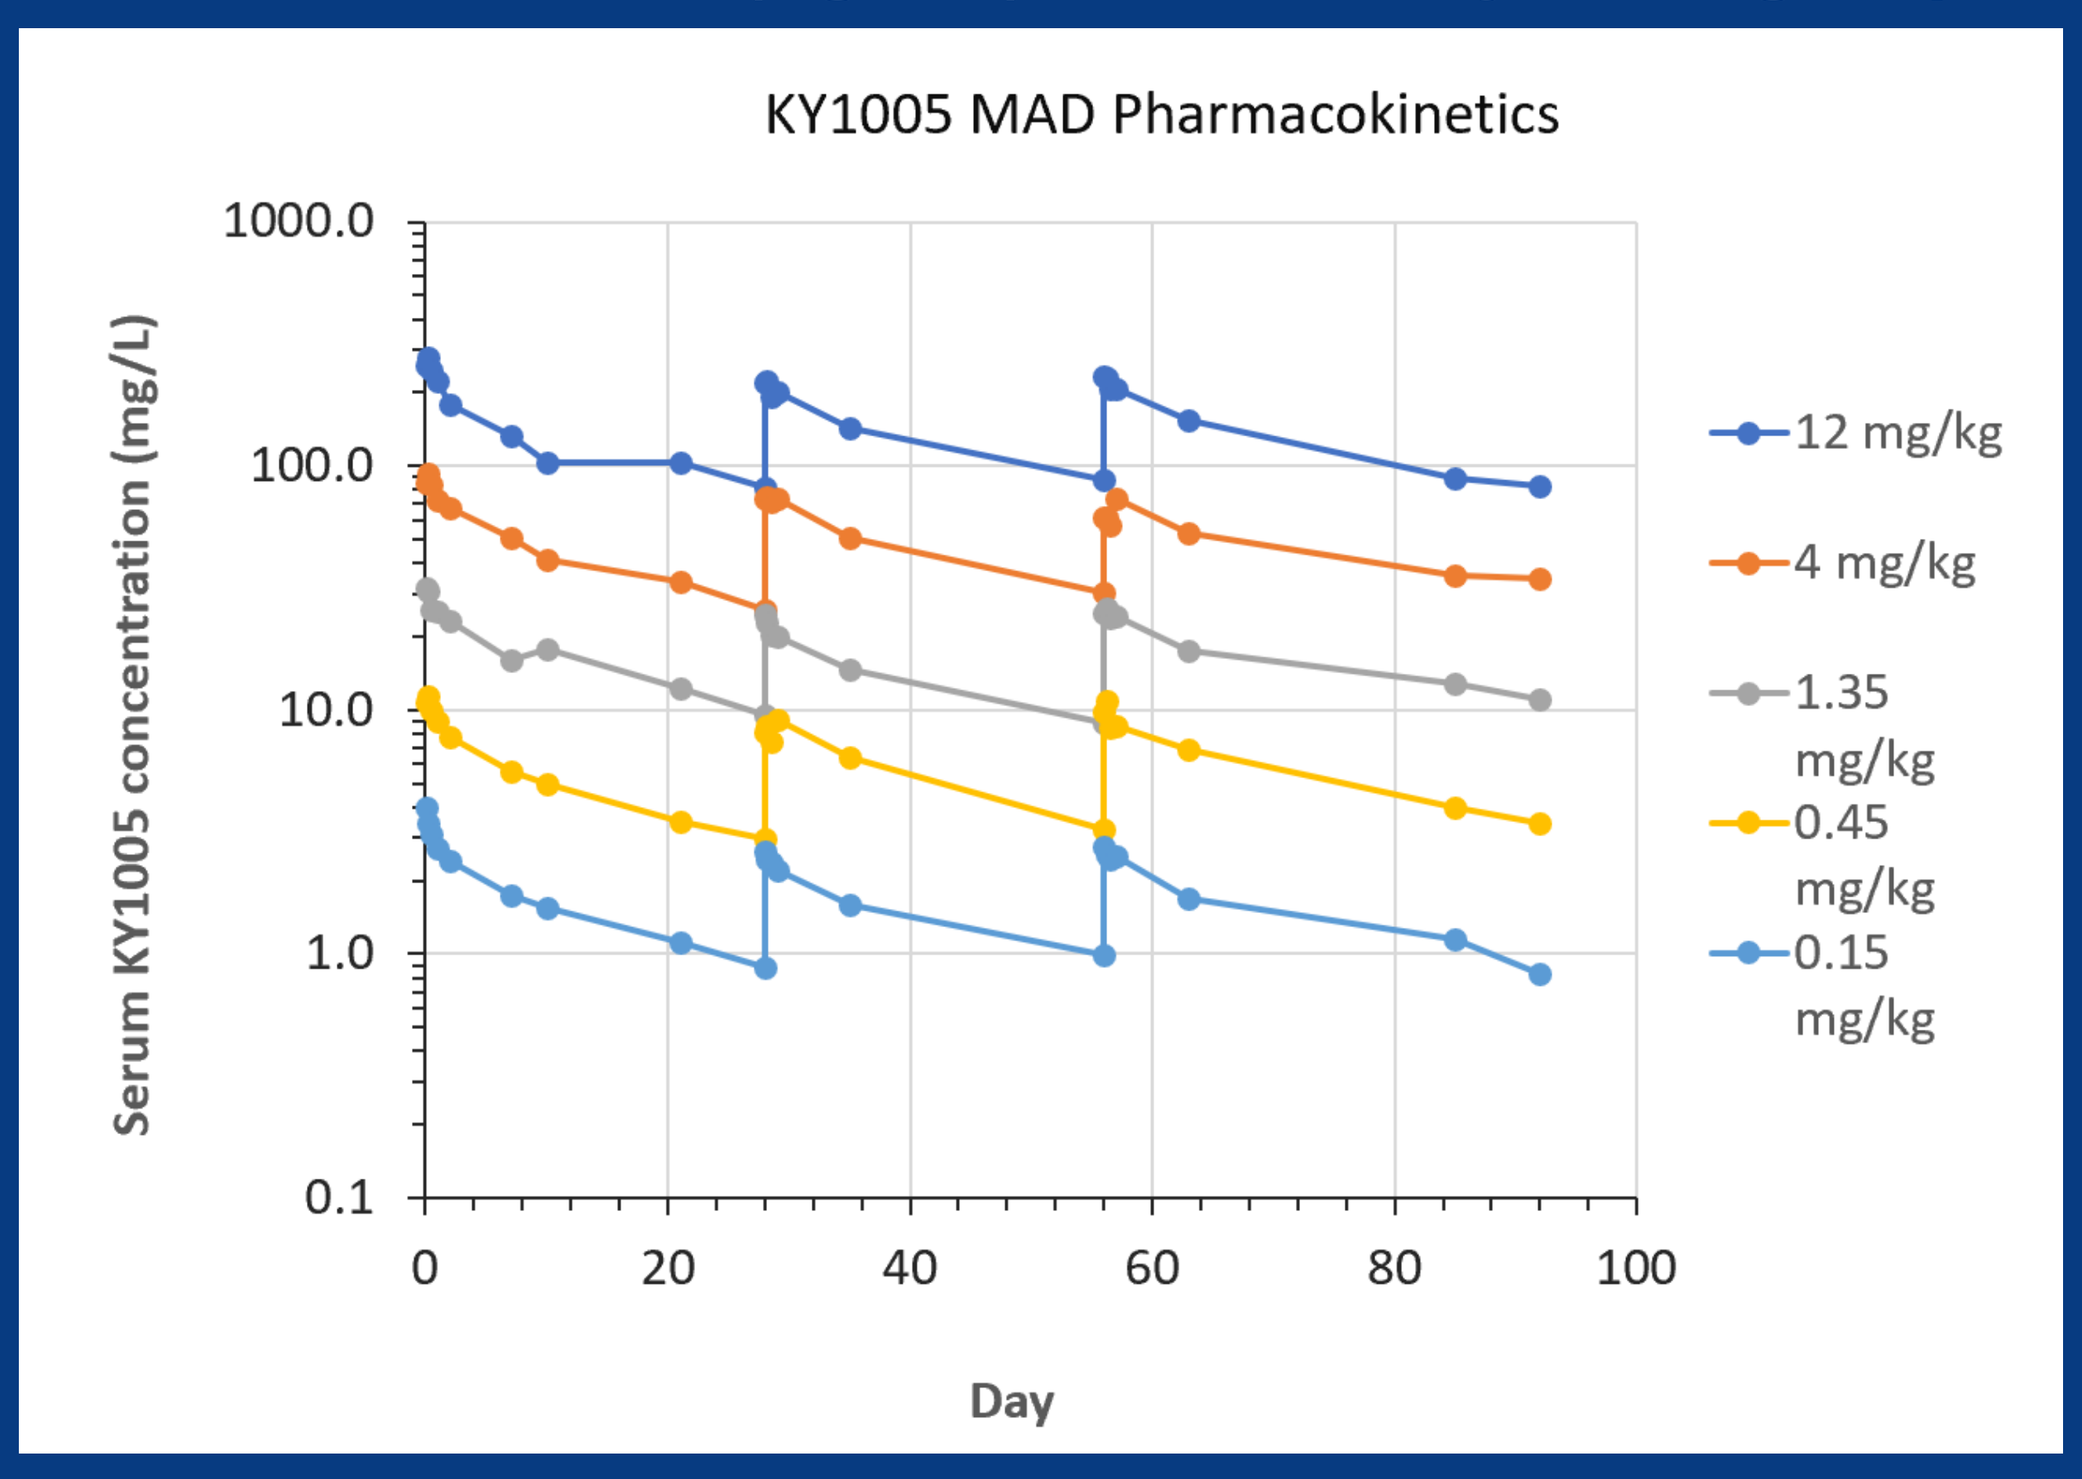
KY1005 MAD Pharmacokinetics
Serum KY1005 concentration (mg/L)
1000.0
100.0
10.0
1.0
0.1
0
20
40
60
80
100
Day
12 mg/kg
4 mg/kg
1.35 mg/kg
0.45 mg/kg
0.15 mg/kg

Conclusions

- KY1005 demonstrated an acceptable safety/tolerability profile.
- Suppression of primary and recall TDAR demonstrates KY1005 is pharmacologically active at doses ≥0.45mg/kg.
- The PK of KY1005 was predictable supporting the use of once monthly dosing.
- These results support the continued development of KY1005 in immune mediated diseases such as AD. A phase 2a study of KY1005 in AD is ongoing (NCT03754309).

Affiliations

1 Centre for Human Drug Research, Leiden, the Netherlands; 2 Kymab Ltd, Cambridge, United Kingdom; 3 Department of Dermatology, Erasmus Medical Centre, Rotterdam, the Netherlands; 4 Leiden Academic Centre for Drug Research, Leiden, the Netherlands; Correspondence; ben.porter-brown@kymab.com Kymab Ltc, Babraham Research Campus, Cambridge, UK CB22 3AT